Poster: 217

Development of Computable Operational Definitions to Maximize Comparability & Consistency Across a Multi-Data Source Global Real-World Effectiveness Program

Aaron WC Kamauu,[1] Craig G Parker,[2] Amanda R Shields,[3] Lisa Glasser,[4] Sabada Dube,[5] Carla Talarico,[6] Sylvia Taylor[5]

[1]Ikaika Health, Bountiful, UT, USA; [2]Navidence LLC, Salt Lake City, UT, USA; [3]Navidence LLC, Chapel Hill, NC, USA; [4]Medical Affairs, Vaccines & Immune Therapies, BioPharmaceuticals Medical, AstraZeneca, Wilmington, DE, USA; [5]Medical Evidence, Vaccine & Immune Therapies, AstraZeneca, Cambridge, UK; [6]Epidemiology, Vaccines & Immune Therapies, BioPharmaceuticals Medical, AstraZeneca, Gaithersburg, MD, USA

Disclosures

- A.K., C.P., and A.S. are partners and owners of Navidence LLC, which is subcontracted via Ikaika Health LLC to provide support to AstraZeneca for various projects and studies
- L.G., S.D., C.T., and S.T. are employees of AstraZeneca

Introduction

- AZD7442, a combination of monoclonal antibodies (tixagevimab/cilgavimab), received emergency use authorization by the FDA in December 2021 for pre-exposure prophylaxis (PrEP) against COVID-19 in patients who are moderately to severely immunocompromised (IC)
- Essential real-world effectiveness assessment is challenged by heterogeneous definitions of IC eligibility, drug distribution differences, inconsistent clinical practices, record-keeping, and availability and completeness of key data across geographic regions/countries and data sources

Objective

- Develop data source-agnostic computable operational definitions (cODs) to support a multinational, multi-data source real-world effectiveness program

Key Take-Home Message

Computable operational definitions support a *multi-data source global* real-world effectiveness program:

- Direct clear and consistent queries across data sources
- Standards-based code lists/value sets
- References for justification of operational definitions and code list mappings (specific to the study element type)
- Context for comparison and interpretation of results across and between data sources

Conclusions

- Every data source is distinct within and across geographic regions/countries, and the unique context is important to the interpretation of results
- Data source-agnostic cODs are foundational to maximize consistency across data sources and countries, comparability of study results, and support reproducibility, even when context may vary
- The authors advise thoughtful creation of clear, consistent, standards-based cODs for all real-world evidence studies, especially those using multiple data sources and/or those submitting evidence to external stakeholders (e.g., regulatory agencies)

Methods

1. Data source-agnostic cODs were established for each of the study elements of the umbrella protocol: eligibility criteria, exposure, baseline characteristics, and outcome measures

Element	Wording	Conceptual Definition	Operational Definition	Related Value Sets
Inclusion 2a1: Active Cancer *Inclusion Criterion*	Source Wording: Active treatment for solid tumor and hematologic malignancies [FDA EUA] Normalized Wording: Active treatment for solid tumor and hematologic malignancies	Diagnosis of cancer AND Treatment for cancer	≥ 1 Diagnosis Record from Any Malignancy, except malignant neoplasm of skin (CCI) Diagnoses (within the 12 months prior to the index date AND any encounter type AND any diagnosis position) OR ≥ 1 Diagnosis Record from Metastatic Solid Tumor (CCI) Diagnoses (within the 12 months prior to the index date AND any encounter type AND any diagnosis position) AND ≥ 1 Medication Record from Cancer Therapies within the 6 months prior to the index date	• Any Malignancy, except malignant neoplasm of skin (CCI) Diagnoses • Metastatic Solid Tumor (CCI) Diagnoses • Cancer Therapies

Label	Type	Extent
Any Malignancy, except malignant neoplasm of skin (CCI) Diagnoses	grouping	0
Any Malignancy, except malignant neoplasm of skin (CCI) Diagnoses (ICD-10-CM)	intensional	1257
Any Malignancy, except malignant neoplasm of skin (CCI) Diagnoses (ICD-9-CM)	intensional	0
Any Malignancy, except malignant neoplasm of skin (CCI) Diagnoses (SNOMED)	extensional	716
Metastatic Solid Tumor (CCI) Diagnoses	grouping	0
Metastatic Solid Tumor (CCI) Diagnoses (ICD-10-CM)	intensional	69
Metastatic Solid Tumor (CCI) Diagnoses (ICD-9-CM)	intensional	0
Metastatic Solid Tumor (CCI) Diagnoses (SNOMED)	extensional	51
Cancer Therapies	grouping	0
Cancer Therapies (ATC)	extensional	263
Cancer Therapies (Name)	extensional	263
Cancer Therapies (RxNorm)	extensional	258

2. The computable nature was facilitated via definition components documented at a granular level in a robust technical platform

Components of a cOD:
- Data variable
- Quantity/magnitude
- Target value, concept, or code list/value set
- Time period qualifier
- Other qualifiers based on the data variable or type of operational definition

Expression to describe how the components relate to each other

3. cODs were tailored to reflect differences for each data source
 – Selected data sources are completing the analysis independently of each other to ensure context and uniqueness of the underlying health system are accounted for

Figure. Leveraging data source-agnostic cODs to create data source-specific cODs

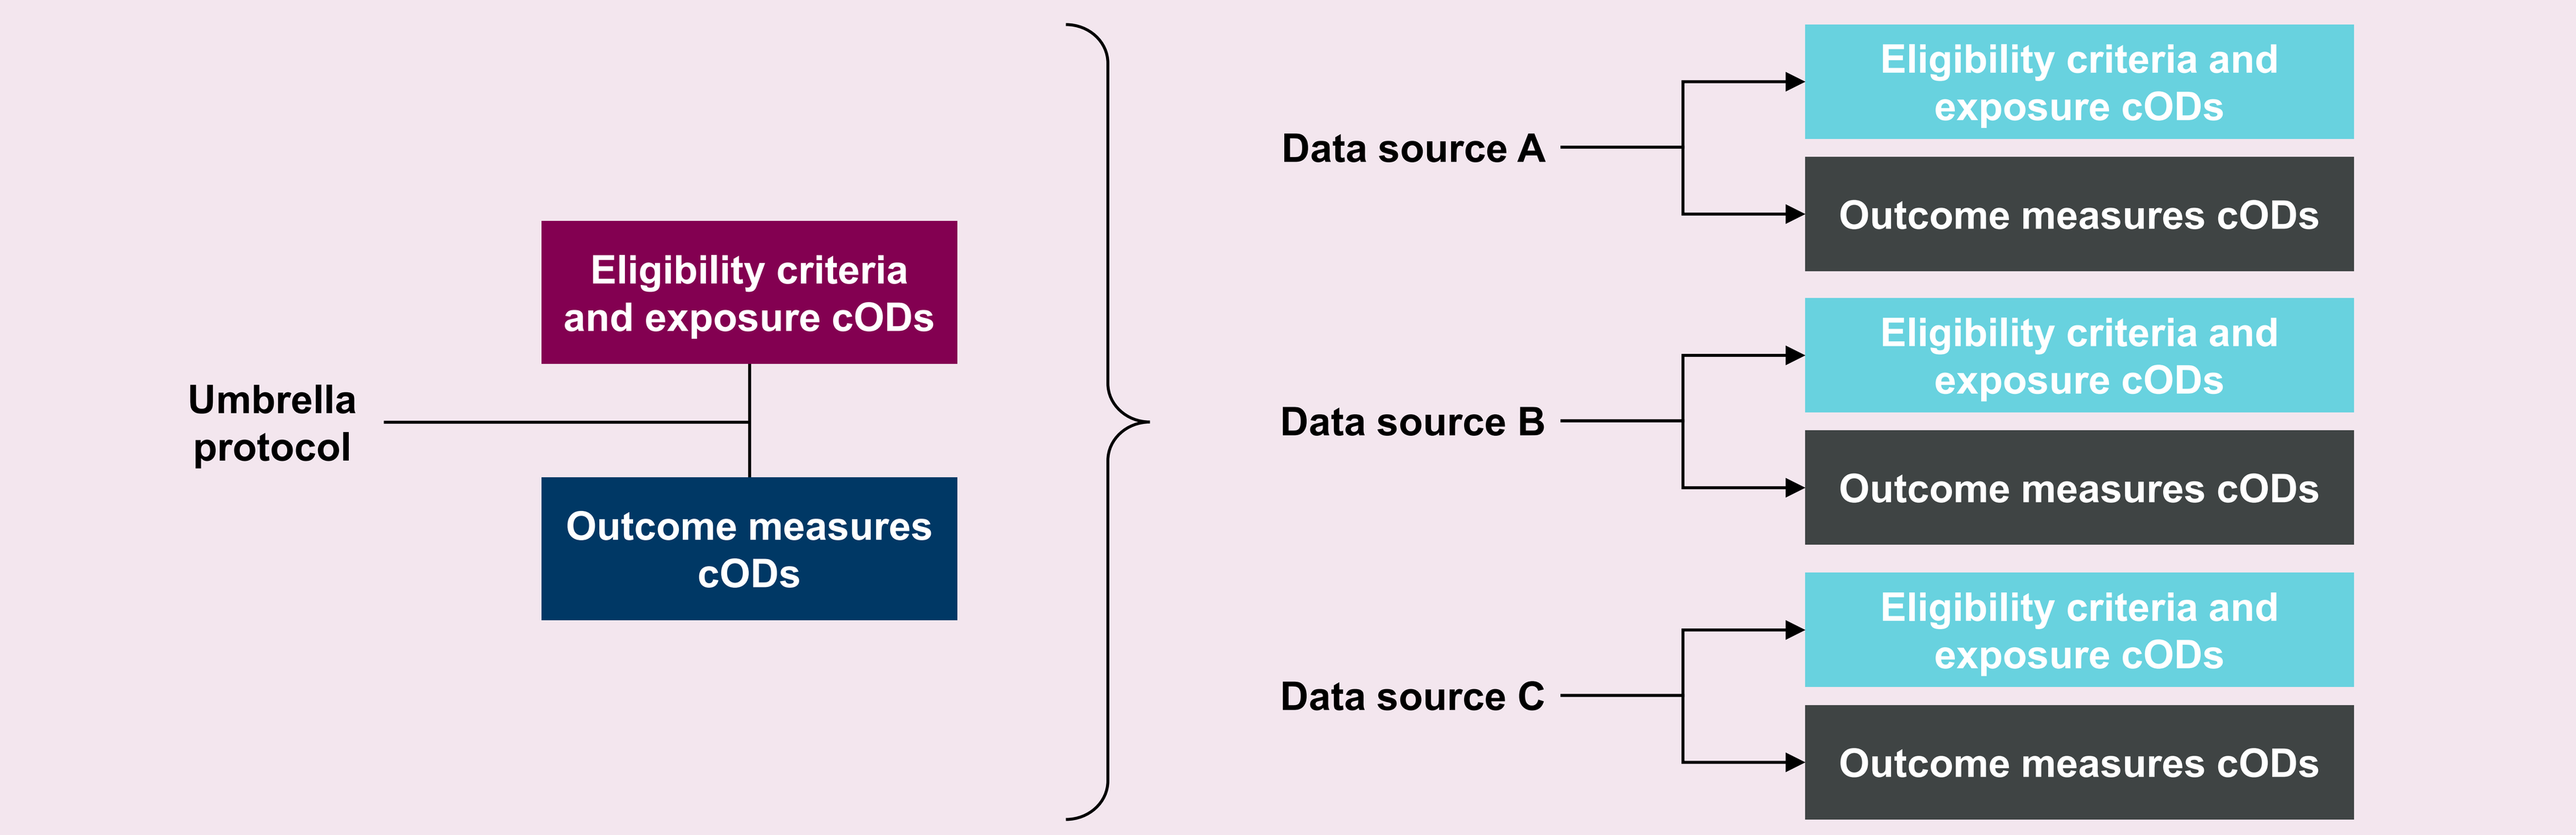

Umbrella protocol
Eligibility criteria and exposure cODs
Outcome measures cODs
Data source A
Data source B
Data source C
Eligibility criteria and exposure cODs
Outcome measures cODs

4. Development of cODs was an iterative process and included:
 – review of literature and published protocols
 – input from epidemiology, statistics, informatics, and medical teams
 – clinical concepts represented by commonly structured data types and standard validated code lists

Results and interpretation

- cODs for 174 study elements were developed for the umbrella study protocol:
 – 17 eligibility criteria
 – 1 exposure
 – 68 baseline characteristics
 – 88 outcomes (for 16 objectives)

- The cODs encompass 38 distinct data variables and 82 standards-based code lists (further delineated by a code system)

Label	Code	Essential
Age	age	True
Date of Birth	date_of_birth	True
Death Record	death_record	True
Encounter Disposition	encounter_disposition	True
Encounter Record	encounter_record	True
Enrollment End Date	enrollment_end_date	True
Enrollment Start Date	enrollment_start_date	True
Sex	sex	True
T-cell helper (CD4) subset panel (Bld)	65758-5	True
Vaccination Record	vaccination_record	True
Admission Date	admission_date	False
Alcohol Use	alcohol_use	False
Area deprivation index	area_deprivation_index	False
Body Mass Index	body_mass_index	False
Cancer Screening Encounter Record	cancer_screening_encounter_record	False
COVID-19 Lab Tests	covid-19_lab_tests	False
Diagnosis Record	diagnosis_record	False
Discharge Date	discharge_date	False
Eligible Dependents	eligible_dependents	False
Immunization record	immunization_record	False
Insurance type	insurance_type	False
Isolation event	isolation_event	False
Marital Status	marital_status	False
Median household salary	median_household_salary	False
Medication Record	medication_record	False

- Direct comparisons across data sources were demonstrated to highlight differences in cODs and their potential impact on the interpretation of analysis results

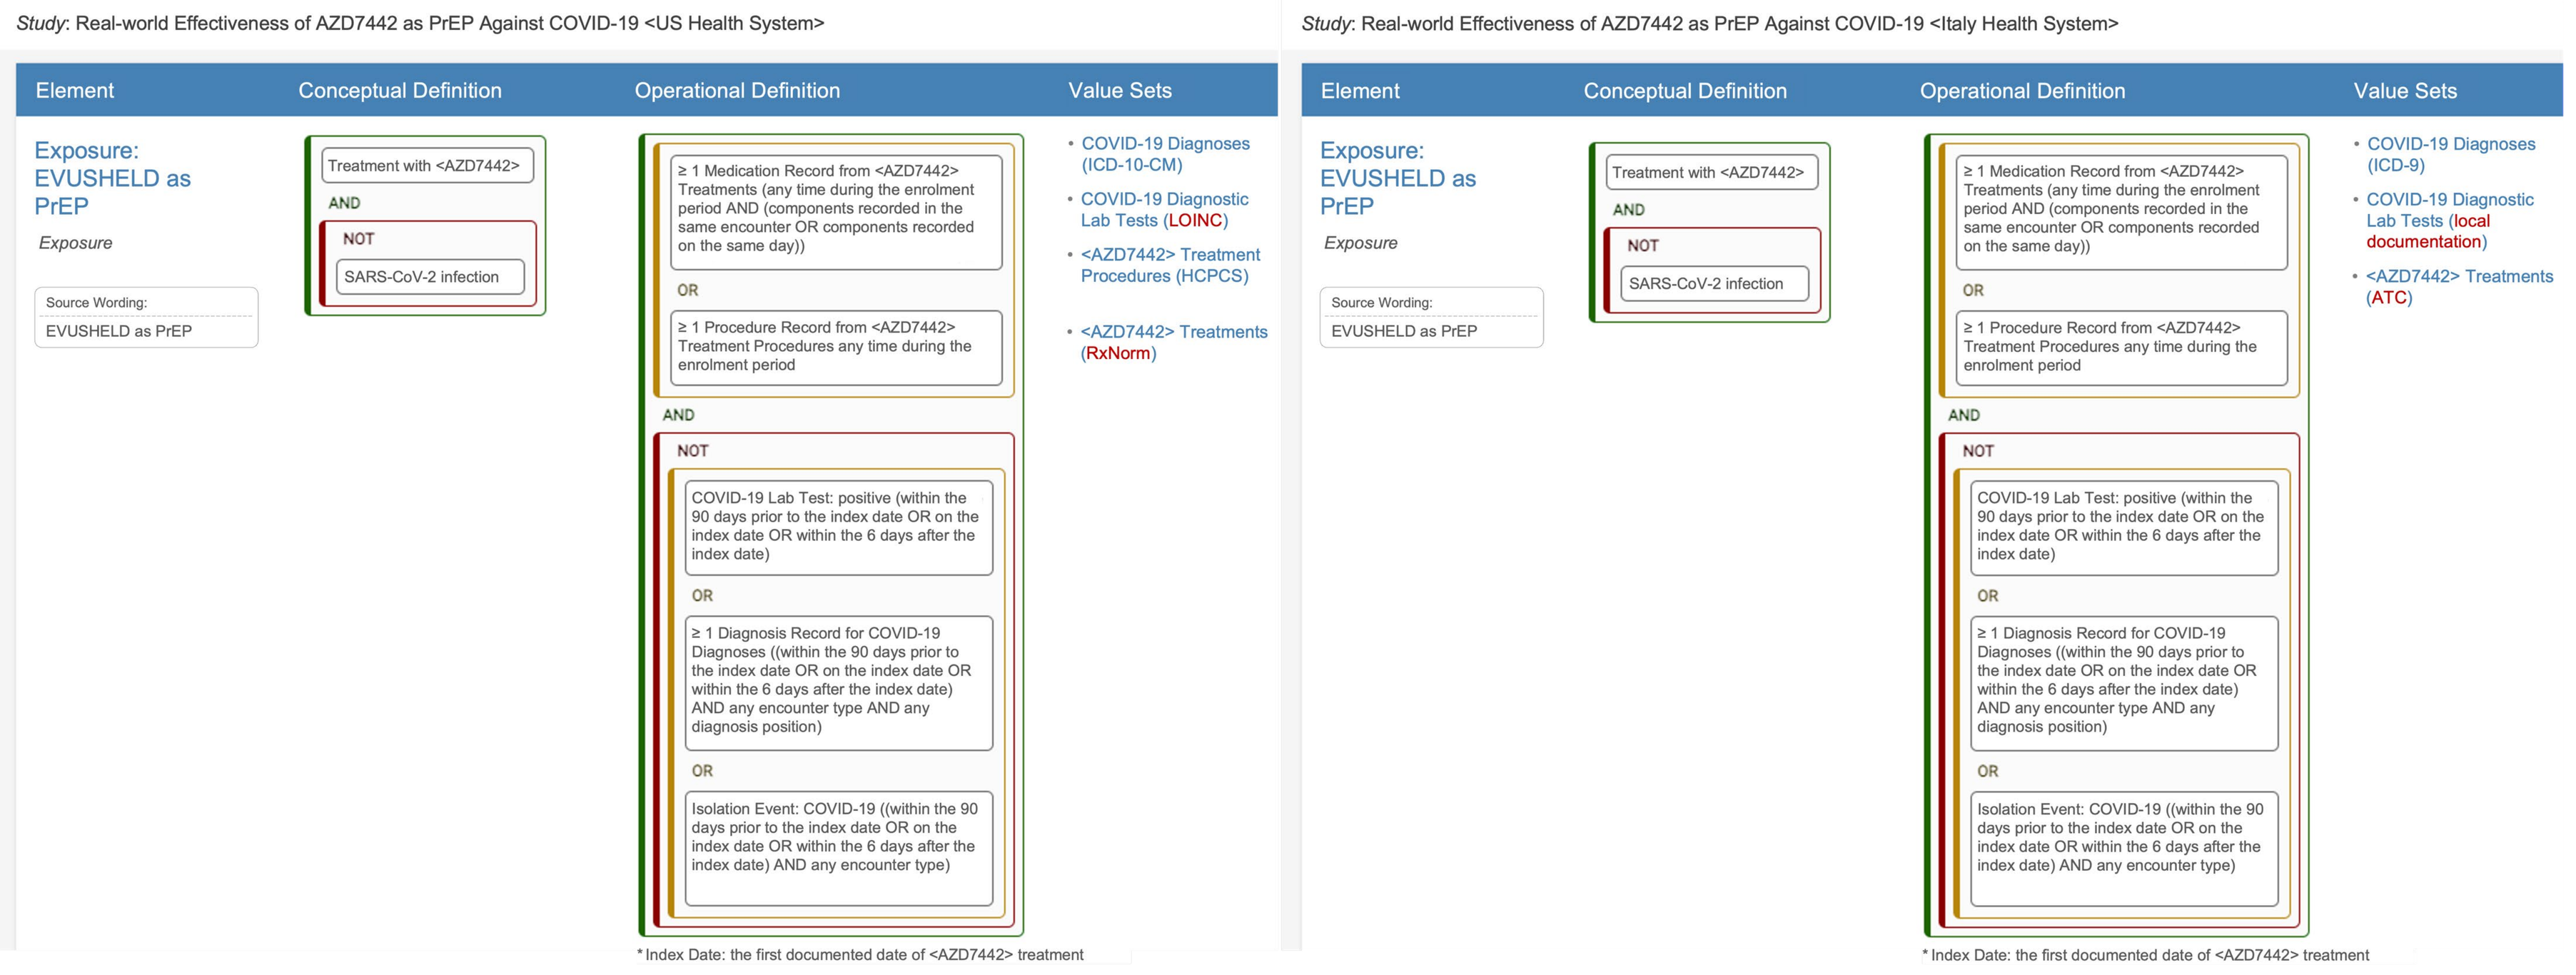

Study: Real-world Effectiveness of AZD7442 as PrEP Against COVID-19 <US Health System>
Element
Conceptual Definition
Operational Definition
Value Sets
Exposure: EVUSHELD as PrEP
Exposure
Source Wording:
EVUSHELD as PrEP
Treatment with <AZD7442>
AND
NOT
SARS-CoV-2 infection
≥ 1 Medication Record from <AZD7442> Treatments (any time during the enrolment period AND (components recorded in the same encounter OR components recorded on the same day))
OR
≥ 1 Procedure Record from <AZD7442> Treatment Procedures any time during the enrolment period
AND
NOT
COVID-19 Lab Test: positive (within the 90 days prior to the index date OR on the index date OR within the 6 days after the index date)
OR
≥ 1 Diagnosis Record for COVID-19 Diagnoses ((within the 90 days prior to the index date OR on the index date OR within the 6 days after the index date) AND any encounter type AND any diagnosis position)
OR
Isolation Event: COVID-19 ((within the 90 days prior to the index date OR on the index date OR within the 6 days after the index date) AND any encounter type)
• COVID-19 Diagnoses (ICD-10-CM)
• COVID-19 Diagnostic Lab Tests (LOINC)
• <AZD7442> Treatment Procedures (HCPCS)
• <AZD7442> Treatments (RxNorm)
* Index Date: the first documented date of <AZD7442> treatment
Study: Real-world Effectiveness of AZD7442 as PrEP Against COVID-19 <Italy Health System>
Element
Conceptual Definition
Operational Definition
Value Sets
Exposure: EVUSHELD as PrEP
Exposure
Source Wording:
EVUSHELD as PrEP
Treatment with <AZD7442>
AND
NOT
SARS-CoV-2 infection
≥ 1 Medication Record from <AZD7442> Treatments (any time during the enrolment period AND (components recorded in the same encounter OR components recorded on the same day))
OR
≥ 1 Procedure Record from <AZD7442> Treatment Procedures any time during the enrolment period
AND
NOT
COVID-19 Lab Test: positive (within the 90 days prior to the index date OR on the index date OR within the 6 days after the index date)
OR
≥ 1 Diagnosis Record for COVID-19 Diagnoses ((within the 90 days prior to the index date OR on the index date OR within the 6 days after the index date) AND any encounter type AND any diagnosis position)
OR
Isolation Event: COVID-19 ((within the 90 days prior to the index date OR on the index date OR within the 6 days after the index date) AND any encounter type)
• COVID-19 Diagnoses (ICD-9)
• COVID-19 Diagnostic Lab Tests (local documentation)
• <AZD7442> Treatments (ATC)
* Index Date: the first documented date of <AZD7442> treatment

- All data sources required some **adaptation**, primarily on coding schemas or definitions unique to the data source
 – Examples of coding schemes include:
 ○ Diagnosis codes: ICD-10-CM, ICD-9, SNOMED
 ○ Procedure codes: ICD-10-PCS, ICD-9, CPT, HCPCS
 ○ Medication codes: RxNorm, ATC-5 (as well as generic and trade names)
 – Examples of unique definitions between data sources:
 ○ IC alignment with national eligibility requirements (e.g., PrEP vs Treatment, IC definitions)
 ○ Setting of AZD7442 administration (e.g., inpatient vs outpatient; IC speciality care vs infectious disease department)
 ○ Patient/population characteristics: socioeconomic status, geographic distribution, common healthcare practices

Limitations

- This program was conducted within the context of a large global real-world effectiveness program covering several data sources across the US, Israel, and specific countries in the EU. Therefore, it does not include countries across all regions of the world, which may limit generalizability of results
 – However, the authors suggest that these methods of using clear, consistent, standards-based cODs would provide greater value for programs conducted across more regions of the world
- While this program resulted in the creation of cODs that cover a variety of medical conditions (either as immunosuppressive conditions for eligibility, many medical conditions as possible comorbidities, or as select effectiveness or safety outcomes), this did not include all disease therapy areas, nor the distinct subtypes of all related medical conditions
 – The authors advise thoughtful creation of clear, consistent, standards-based cODs for medical conditions that are relevant to individual clinical research needs and purpose
 – In addition, the authors advise leveraging cODs with the appropriate context of the study element. For example, different cODs may be appropriate for the same clinical condition/conceptual definition, when applied as an inclusion criterion versus a baseline comorbidity versus an outcome measure

Supplementary content

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of this poster. Alternatively, please click on the link below:
https://bit.ly/3Qiyuyv

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission.

Acknowledgments

This project was supported by AstraZeneca. The authors would like to acknowledge Megan Somerday and Allise Kamauu for their contributions in support of this project.

Abbreviations and codes

ATC-5, Anatomical Therapeutic Chemical 5th level; CCI, Charlson Comorbidity Index; CM, Clinical Modification; cOD, computable operational definition; COVID-19, coronavirus disease 2019; CPT, Current Procedural Terminology; EUA, Emergency Use Authorization; FDA, The US Food and Drug Administration HCPCS, Healthcare Common Procedure Coding System; IC, immunocompromised; ICD-9, International Classification of Diseases, 9th Revision; ICD-10, International Classification of Diseases, 10th Revision; LOINC, Logical Observation Identifiers Names and Codes; PCR, polymerase chain reaction; PCS, Procedure Coding System; PrEP, pre-exposure prophylaxis; RxNorm, standardized nomenclature for clinical drugs; SARS-CoV-2, severe acute respiratory syndrome coronavirus 2; SNOMED, Systematized Nomenclature of Medicine.

Presented at the 39th International Conference on Pharmacoepidemiology & Therapeutic Risk Management (ICPE) | August 23–27, 2023, Halifax, Nova Scotia, Canada